Presented at the 2024 European Society for Medical Oncology Congress • 13-17 September 2024 • Barcelona, Spain

1136P

Cosibelimab in Advanced Cutaneous Squamous Cell Carcinoma (CSCC): Longer-term Efficacy and Safety Results From Pivotal Study

E. Muñoz-Couselo*,[1] H. Montaudié,[2] M.A. Berciano-Guerrero,[3] M.D.C. Alamo De La Gala,[4] J. Charles,[5] S. Trabelsi,[5] G. Quereux,[6] C. Nardin,[7] R. Yaya Tur,[8] S. Dalle,[9] M. Beylot-Barry,[10] R. Ladwa,[11] M. McGrath,[12] D. Brungs,[13] D. Harris,[14] H. Shue,[15] A. Tazbirkova,[16] S. Fourie,[17] D. Malan,[18] J. Oliviero,[19] P. Clingan[13]

[1]Medical Oncology, Vall d'Hebron Hospital & Vall d'Hebron Institute of Oncology (VHIO), Barcelona, Spain; [2]Dermatology, Centre Hospitalier Universitaire de Nice, Université Côte d'Azur, Nice, France; [3]Medical Oncology Department, Regional University Hospital. IBIMA Plataforma BIONAND, Malaga, Spain; [4]Medical Oncology, Hospital Universitario Virgen Macarena, Seville, Spain; [5]Dermatology, Grenoble Alpes University Hospital/Grenoble Alpes University UGA, Grenoble, France; [6]Dermatology, Nantes University Hospital, Nantes, France; [7]Dermatology, Centre Hospitalier Universitaire de Besancon, Besancon, France; [8]Medical Oncology, Fundacion Instituto Valenciano de Oncologia, Valencia, Spain; [9]Dermatology, Hospices Civils de Lyon, Pierre Benite, France; [10]Dermatology, CHU Bordeaux - Hopital St. André, Bordeaux, France; [11]Division of Cancer Services, Princess Alexandra Hospital, Woolloongabba, QLD, Australia; [12]Medical Oncology, Gallipoli Medical Research Foundation, Greenslopes Private Hospital, Brisbane, QLD, Australia; [13]Medical Oncology, Cancer Care Wollongong, Wollongong, NSW, Australia; [14]Oncology, Christchurch Hospital, Te Whatu Ora, Waitaha/Canterbury, Christchurch, New Zealand; [15]University of The Sunshine Coast Clinical Trials Unit, Sunshine Coast Hematology and Oncology Clinic, Buderim, QLD, Australia; [16]Medical Oncology, Pindara Private Hospital, Benowa, QLD, Australia; [17]Oncology, Wilgers Oncology Center, Pretoria, South Africa; [18]Research Unit, Phoenix Pharma (Pty) Ltd, Port Elizabeth, South Africa; [19]Executive, Checkpoint Therapeutics, Waltham, MA, United States of America

OBJECTIVES

- **The primary objective of the open-label, multicenter, multiregional, multicohort, pivotal phase 1 trial was to evaluate the objective response rate (ORR; complete response + partial response) by independent central review (ICR) per Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST 1.1; for scans) and clinical response criteria using bidimensional measurements in accordance with World Health Organization (WHO) criteria (for photos) assessed by CSCC group (ClinicalTrials.gov identifier: NCT03212404)**
- **Key secondary objectives included evaluation of duration of response (DOR) by ICR and assessment of safety and tolerability**
- **Here, we present longer-term follow-up efficacy and safety data from the pivotal phase 1 trial of cosibelimab in patients with metastatic CSCC (mCSCC) or locally advanced CSCC (laCSCC) (collectively, advanced CSCC)**

CONCLUSIONS

- **Cosibelimab demonstrated robust and durable ORRs and complete response rates in patients with advanced CSCC**
 - **ORRs of 50.0% and 54.8% were achieved in mCSCC and laCSCC, respectively**
 - **Results demonstrate a deepening of response over time, resulting in higher complete response rates than initially reported at the primary analysis. Overall, the complete response rates are 12.8% and 25.8% in mCSCC and laCSCC, respectively**
 - **A clinically meaningful duration of response (DOR) also was observed with cosibelimab, with medians not yet reached**
- **Overall, a manageable safety profile, regardless of attribution, was observed, with notable low rates of overall treatment-emergent adverse events (TEAEs), severe immune-related adverse events (irAEs), and treatment discontinuations**

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without written permission of the authors.

*Presenting Author: E. Muñoz-Couselo; emunoz@vhio.net

Declaration of Interests: EMC has served as a consultant and/or scientific advisor for Bristol Myers Squibb, Merck & Co., Novartis, Pierre Fabre, Sanofi, Immunocore, Regeneron, Menarini, and Roche; and has served as a clinical investigator for the CK-301-101 trial sponsored by Checkpoint Therapeutics.

Introduction

- Cutaneous squamous cell carcinoma (CSCC) is the second most common form of skin cancer, and advanced or metastatic disease is associated with substantial morbidity and mortality[1]
 - In a 2020 study from the Netherlands, the authors estimated that European CSCC incidence rates would increase by 23.0% for males and 29.4% for females between 2017 and 2027[2]
 - Similarly, the incidence of CSCC is increasing in the United States, United Kingdom, and Australia[3-5]
 - This increase in incidence will be associated with corresponding increases in morbidity, mortality, public health burden, and social costs[1]
- Programmed death receptor-1 (PD-1)–blocking antibodies are approved as monotherapy for patients with advanced CSCC (metastatic [mCSCC] or locally advanced [laCSCC]) who are not candidates for curative surgery or radiation[6]
- Cosibelimab is a high-affinity, fully human monoclonal antibody that binds to programmed death-ligand 1 (PD-L1) and inhibits its interaction with the PD-1 and B7.1 receptors. Additionally, cosibelimab has a functional fragment crystallizable (Fc) domain capable of inducing antibody-dependent cellular cytotoxicity (ADCC) and complement-dependent cytotoxicity (CDC) against tumor cells (Figure 1)[7]

Figure 1. Cosibelimab dual mechanisms of action (MOAs). (A) Modeling predicts that cosibelimab sustains >99% tumor target occupancy to block PD-L1 interaction with PD-1 and reactivate T cells.[7,8] (B) Cosibelimab contains a functional Fc domain capable of inducing ADCC and CDC against tumor cells.[7]

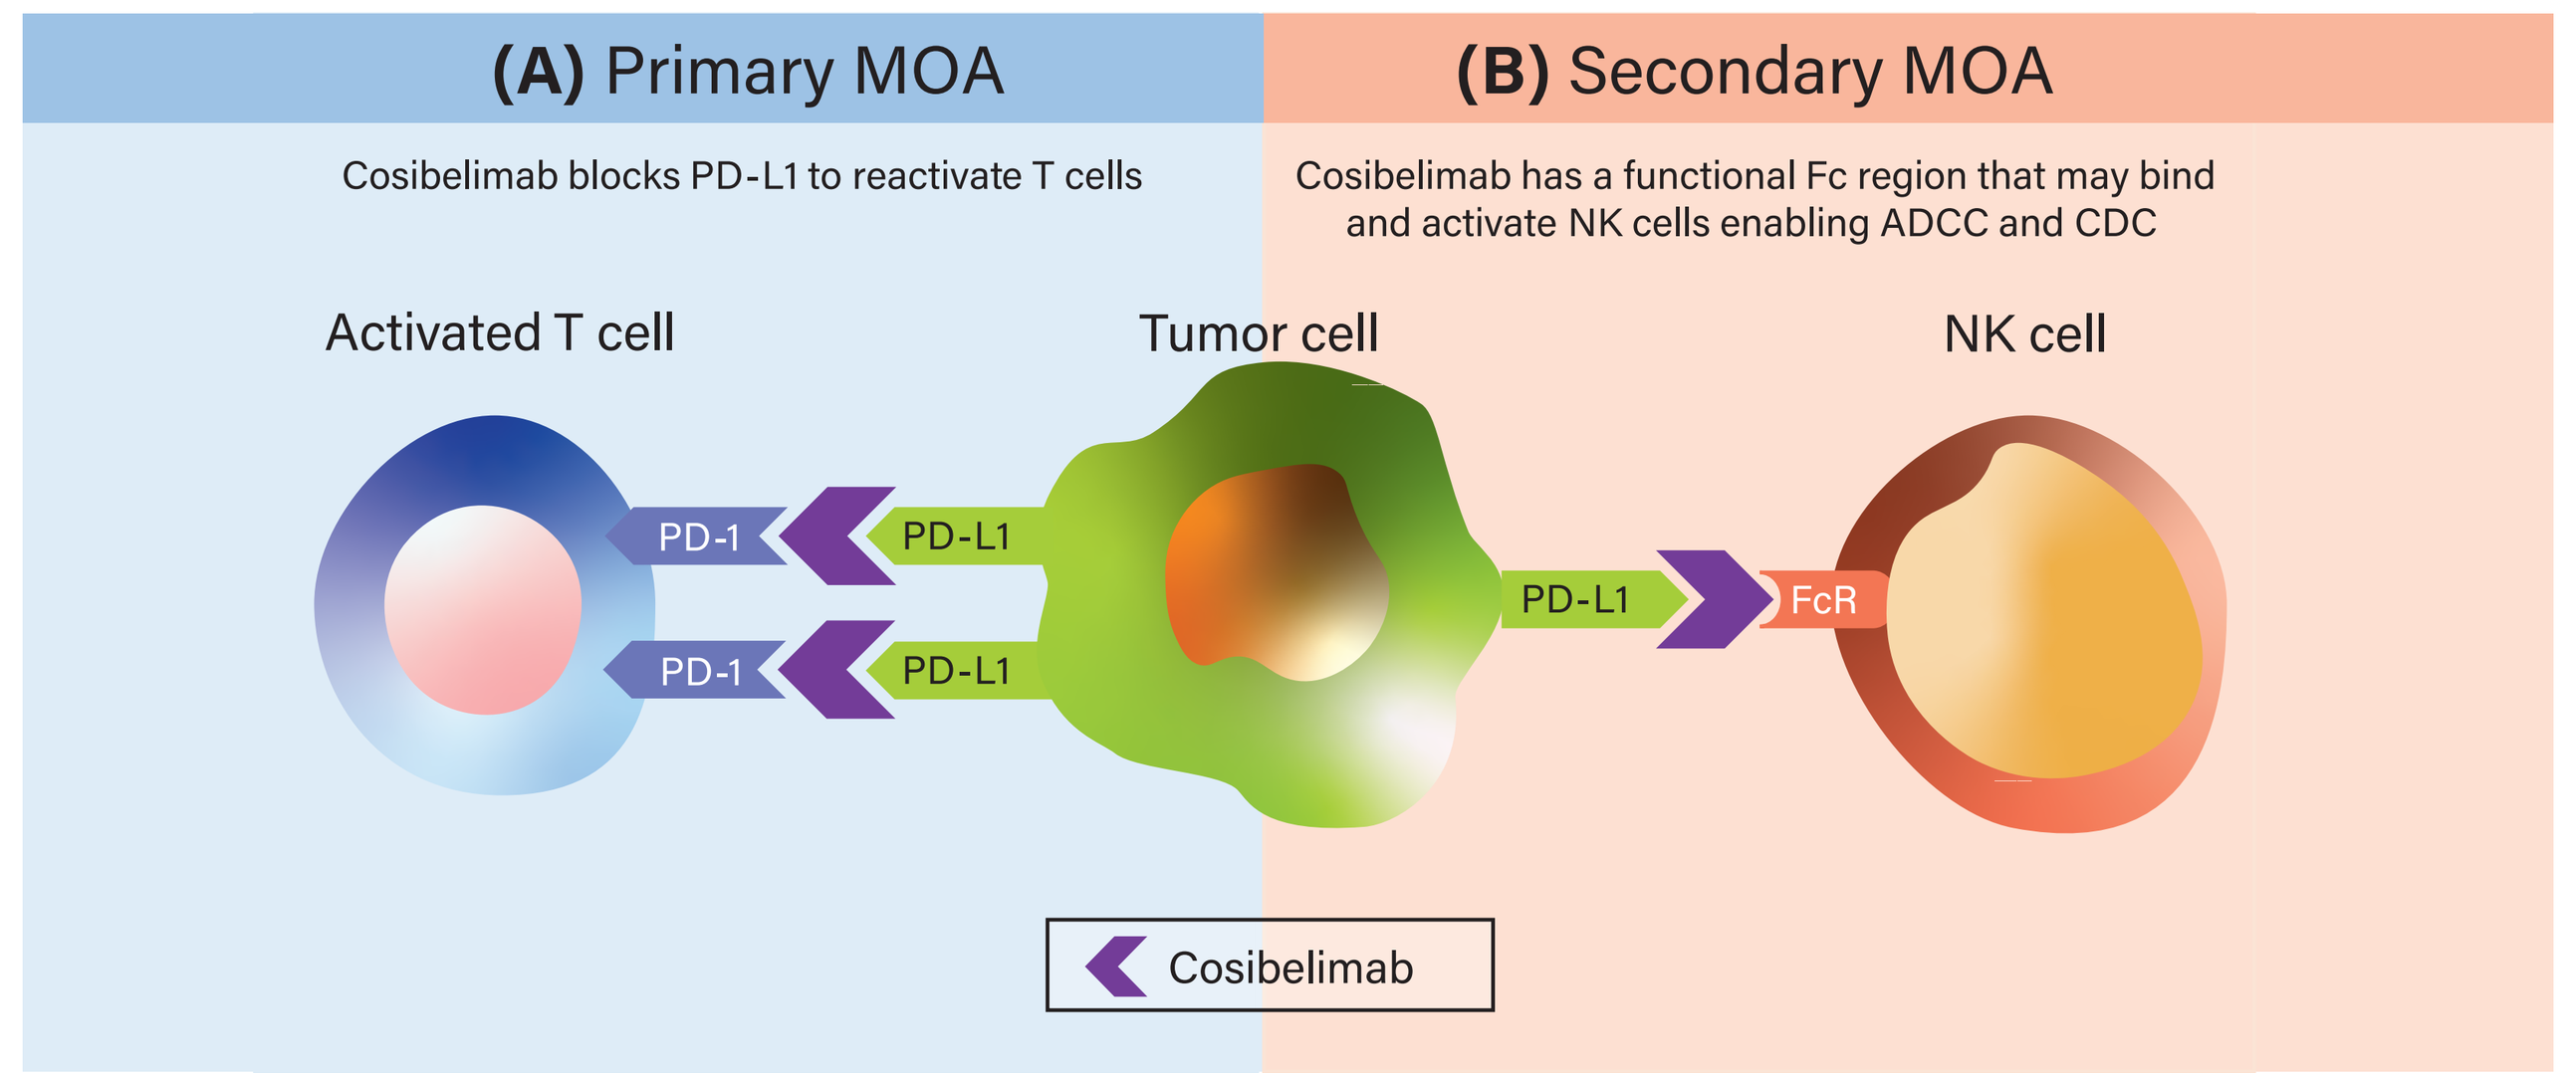

(A) Primary MOA
Cosibelimab blocks PD-L1 to reactivate T cells
(B) Secondary MOA
Cosibelimab has a functional Fc region that may bind and activate NK cells enabling ADCC and CDC
Activated T cell
Tumor cell
NK cell
PD-1
PD-L1
PD-1
PD-L1
PD-L1
FcR
Cosibelimab

ADCC, antibody-dependent cellular cytotoxicity; CDC, complement-dependent cytotoxicity; Fc, fragment crystallizable; FcR, Fc receptor; MOA, mechanism of action; NK, natural killer; PD-1, programmed death receptor-1; PD-L1, programmed death-ligand 1.

Methods

- Key eligibility criteria for patients included in the study are shown in Table 1

Table 1. Key Eligibility Criteria

Inclusion	Exclusion
Age ≥18 years	Prior immune checkpoint inhibitor therapy
ECOG PS 0 or 1 and life expectancy ≥3 months	Active, suspected, or documented history of autoimmune disease
Histologically confirmed diagnosis of mCSCC (nodal and/or distant metastatic disease) or unresectable laCSCC (without nodal or distant metastatic disease) not amenable to local therapy	Concurrent immunosuppressive doses of steroids (>10 mg/day prednisone or equivalent)

ECOG PS, Eastern Cooperative Oncology Group performance status; laCSCC, locally advanced cutaneous squamous cell carcinoma; mCSCC, metastatic cutaneous squamous cell carcinoma.

- Patients received intravenous cosibelimab 800 mg every 2 weeks (Q2W; Group 1, mCSCC; Group 2, laCSCC) or 1200 mg every 3 weeks (Q3W; Group 3, mCSCC); treatment was continued until confirmed complete response, worsening disease progression, toxicity, or clinical deterioration followed by post-treatment follow-up (Figure 2)
 - Cosibelimab dosing regimens of 800 mg Q2W and 1200 mg Q3W are comparable based on the pharmacokinetics-related criteria outlined in the US Food and Drug Administration guidance for supporting alternative dosing regimens for PD-L1–blocking antibodies[9,10]
- Patients were contacted by telephone quarterly for survival and tumor treatment status after treatment was stopped and the necessary follow-up visits were completed

Figure 2. Study design for patients with mCSCC or laCSCC who received cosibelimab Q2W or Q3W.

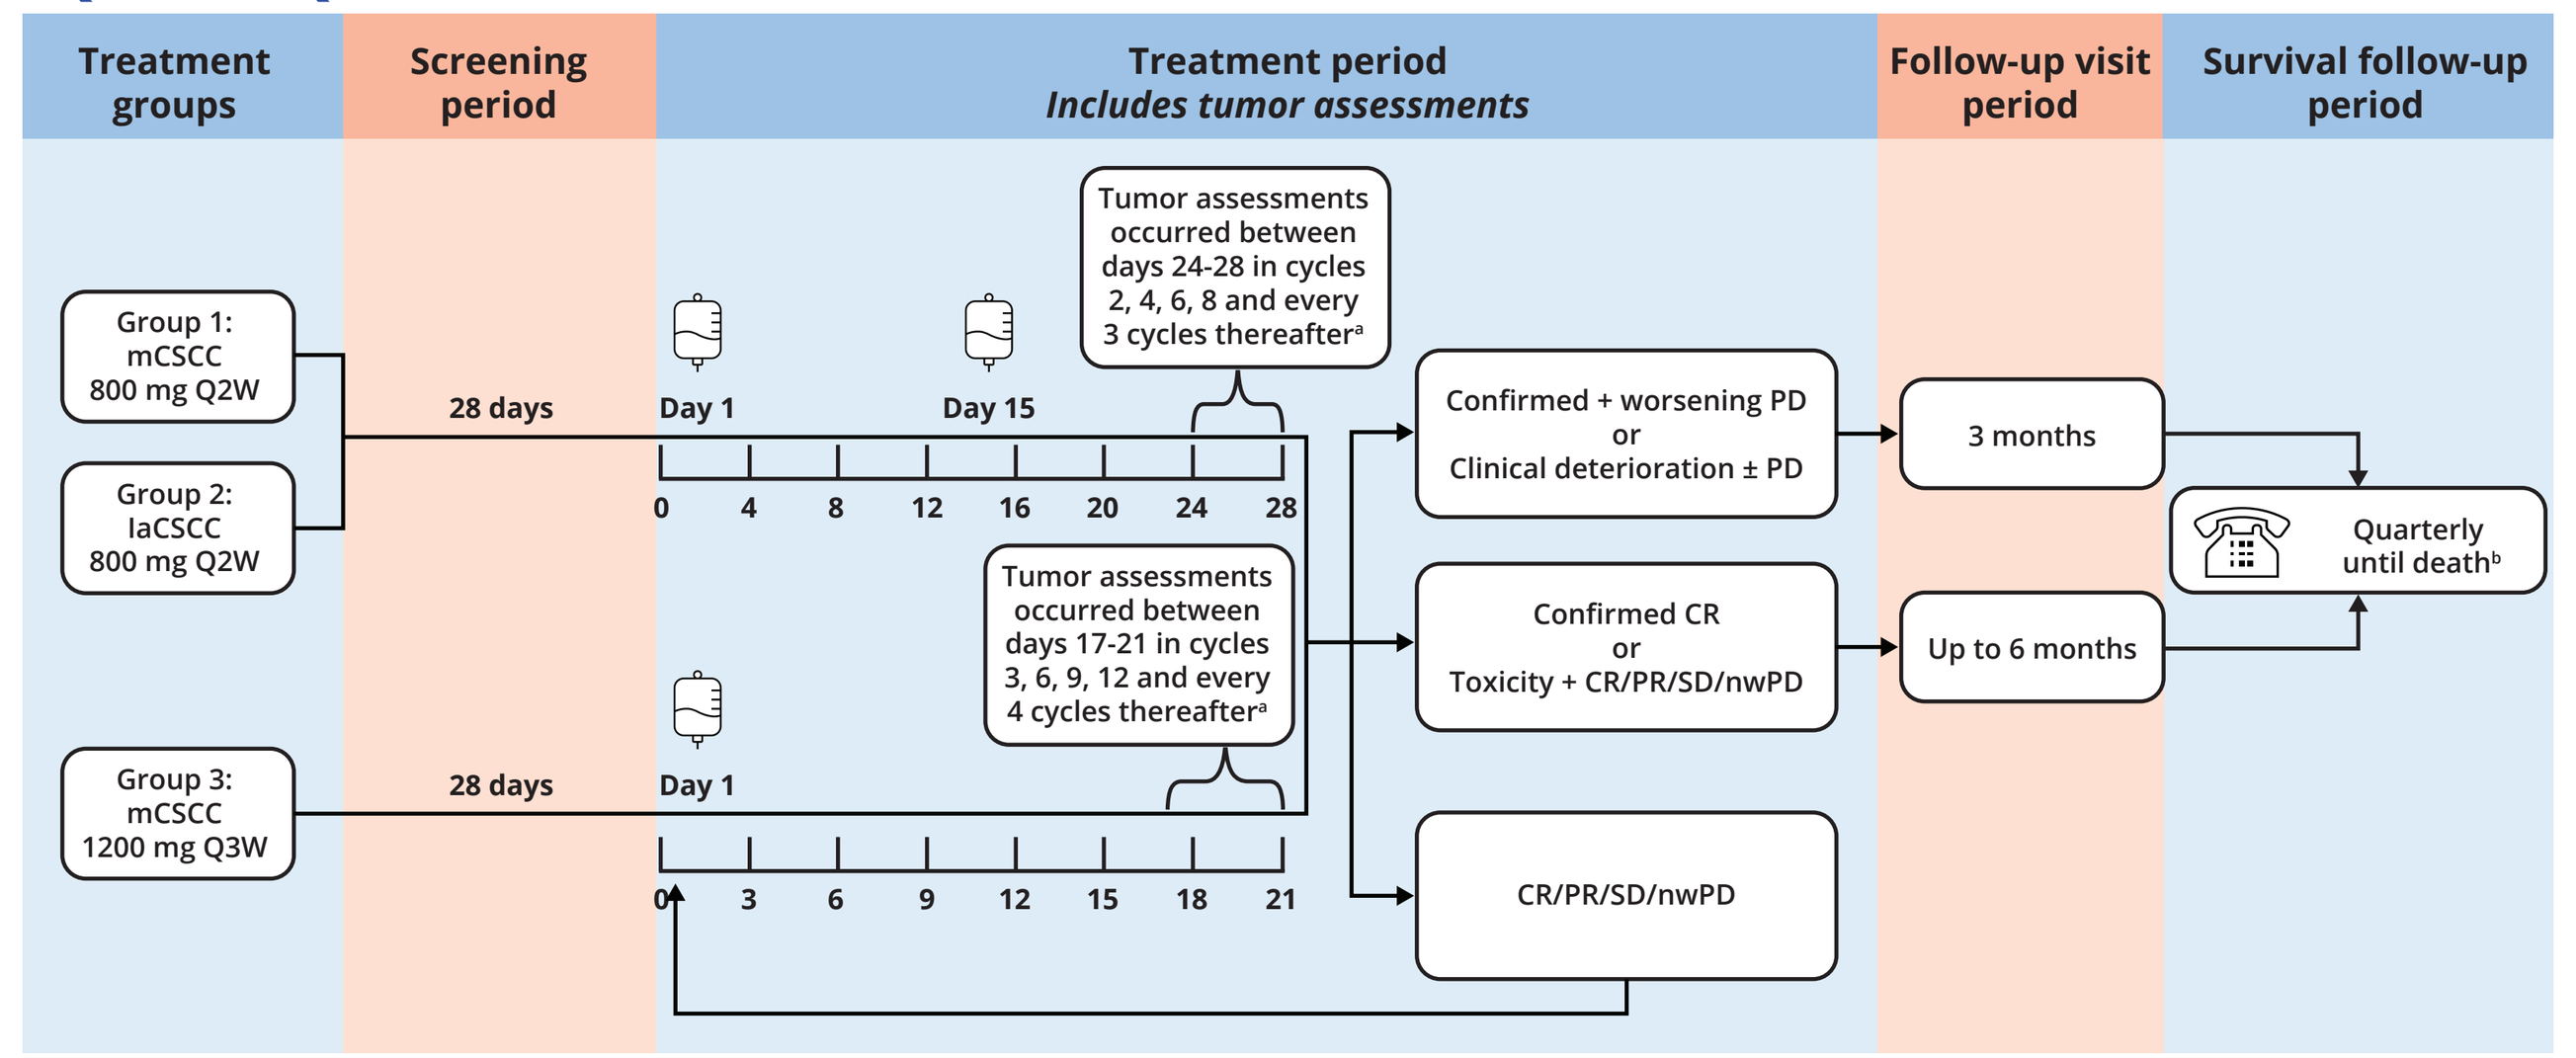

Treatment groups
Screening period
Treatment period
Includes tumor assessments
Follow-up visit period
Survival follow-up period
Group 1: mCSCC 800 mg Q2W
Group 2: laCSCC 800 mg Q2W
Group 3: mCSCC 1200 mg Q3W
28 days
28 days
Day 1
Day 15
Day 1
0 4 8 12 16 20 24 28
0 3 6 9 12 15 18 21
Tumor assessments occurred between days 24-28 in cycles 2, 4, 6, 8 and every 3 cycles thereafter[a]
Tumor assessments occurred between days 17-21 in cycles 3, 6, 9, 12 and every 4 cycles thereafter[a]
Confirmed + worsening PD or Clinical deterioration ± PD
Confirmed CR or Toxicity + CR/PR/SD/nwPD
CR/PR/SD/nwPD
3 months
Up to 6 months
Quarterly until death[b]

CR, complete response; laCSCC, locally advanced cutaneous squamous cell carcinoma; mCSCC, metastatic cutaneous squamous cell carcinoma; nwPD, nonworsening progressive disease; PD, progressive disease; PR, partial response; Q2W, every 2 weeks; Q3W, every 3 weeks; SD, stable disease. [a]End-of-cycle tumor assessments informed the decision to treat patients with additional cycles or begin follow-up visits. [b]After patients stop treatment and complete the necessary follow-up visits, they are contacted by telephone quarterly for survival and tumor treatment status, if available, until death.

Results

Baseline Characteristics

- As of 31 March 2023, 192 patients were enrolled and dosed in the 3 CSCC groups (Group 1, n=78; Group 2, n=58; Group 3, n=56) and considered as the safety population
- Patients eligible for long-term efficacy assessment in Groups 1 and 2 (Group 1, n=78; Group 2, n=31) were considered as the efficacy population (Table 2)
 - Represents all patients included in the prespecified primary analysis as outlined in the statistical analysis plan (primary analysis data cutoff of 18 November 2021)

Table 2. Patient Demographics and Baseline Characteristics

Demographic/Characteristic, n (%)	Group 1 mCSCC (n=78)	Group 2 laCSCC (n=31)
Sex		
Female	19 (24.4)	12 (38.7)
Male	59 (75.6)	19 (61.3)
Median age		
<65 years	22 (28.2)	2 (6.5)
≥65 years	56 (71.8)	29 (93.5)
Race		
White	69 (88.5)	24 (77.4)
Asian	6 (7.7)	0
Black or African American	1 (1.3)	0
Other	0	1 (3.2)
Missing[a]	2 (2.6)	6 (19.4)
Ethnicity		
Not Hispanic or Latino	73 (93.6)	19 (61.3)
Hispanic or Latino	3 (3.8)	5 (16.1)
Unknown	0	1 (3.2)
Missing[a]	2 (2.6)	6 (19.4)
Country/Region		
Asia	6 (7.7)	0
Australia/New Zealand	45 (57.7)	13 (41.9)
Europe	19 (24.4)	17 (54.8)
South Africa	8 (10.3)	1 (3.2)
ECOG PS		
0	23 (29.5)	14 (45.2)
1	55 (70.5)	17 (54.8)
Primary CSCC site		
Head/Neck	46 (59.0)	28 (90.3)
Extremity	18 (23.1)	2 (6.5)
Trunk	9 (11.5)	1 (3.2)
Other	5 (6.4)	0
Type of metastatic disease		
Distant	52 (66.7)	NA
Nodal only	26 (33.3)	NA
Prior cancer-related surgery		
Yes	47 (60.3)	25 (80.6)
No	31 (39.7)	6 (19.4)
Prior radiation therapy		
Yes	51 (65.4)	24 (77.4)
No	27 (34.6)	7 (22.6)
Prior systemic therapy		
Yes	7 (9.0)	1 (3.2)
No	71 (91.0)	30 (96.8)

CSCC, cutaneous squamous cell carcinoma; ECOG PS, Eastern Cooperative Oncology Group performance status; laCSCC, locally advanced CSCC; mCSCC, metastatic CSCC; NA, not applicable. [a]Some sites in Europe did not report race and ethnicity owing to privacy laws.

Efficacy

- With a median follow-up duration of 29.3 months (range, 0.4-52.0 months) for Group 1 and 24.1 months (range, 2.8-37.3 months) for Group 2, the ORR per ICR was 50.0% (95% confidence interval [CI], 38.5%-61.5%) and 54.8% (95% CI, 36.0%-72.7%), respectively (Table 3)
- The complete response rate was 12.8% and 25.8% in Group 1 (mCSCC) and Group 2 (laCSCC), respectively
- ORRs and complete response rates at the primary analysis and 16 months of additional follow-up are shown in Figure 3
- Median duration of response (DOR) has not been reached in either group, with a probability of maintaining response at 24 months of 72.1% and 80.2% for Groups 1 and 2, respectively (Figure 4)

Table 3. Tumor Response to Cosibelimab by ICR

	Group 1 mCSCC (n=78)	Group 2 laCSCC (n=31)
Median follow-up duration (95% CI), months	29.3 (25.3-34.8)	24.1 (22.6-26.9)
ORR (95% CI), %	50.0 (38.5-61.5)	54.8 (36.0-72.7)
Best overall response, n (%)		
Complete response	10 (12.8)	8 (25.8)
Partial response	29 (37.2)	9 (29.0)
Stable disease	11 (14.1)	10 (32.3)
Progressive disease	20 (25.6)	3 (9.7)
Not evaluable	8 (10.3)	1 (3.2)
Median observed time to response (range), months	1.9 (1.6-16.9)	3.6 (1.7-10.1)
Median DOR (range), months	NR (1.4-45.3)	NR (8.3-31.3)
KM-estimated 6-month DOR probability (95% CI), %	89.5 (74.3-95.9)	100 (NE)
KM-estimated 12-month DOR probability (95% CI), %	75.4 (57.9-86.4)	88.2 (60.6-96.9)
KM-estimated 24-month DOR probability (95% CI), %	72.1 (54.1-84.0)	80.2 (49.6-93.3)

CI, confidence interval; DOR, duration of response; ICR, independent central review; KM, Kaplan-Meier; laCSCC, locally advanced cutaneous squamous cell carcinoma; mCSCC, metastatic cutaneous squamous cell carcinoma; NE, not evaluable; NR, not reached; ORR, objective response rate.

Figure 3. (A) ORRs and (B) complete response rates per ICR.

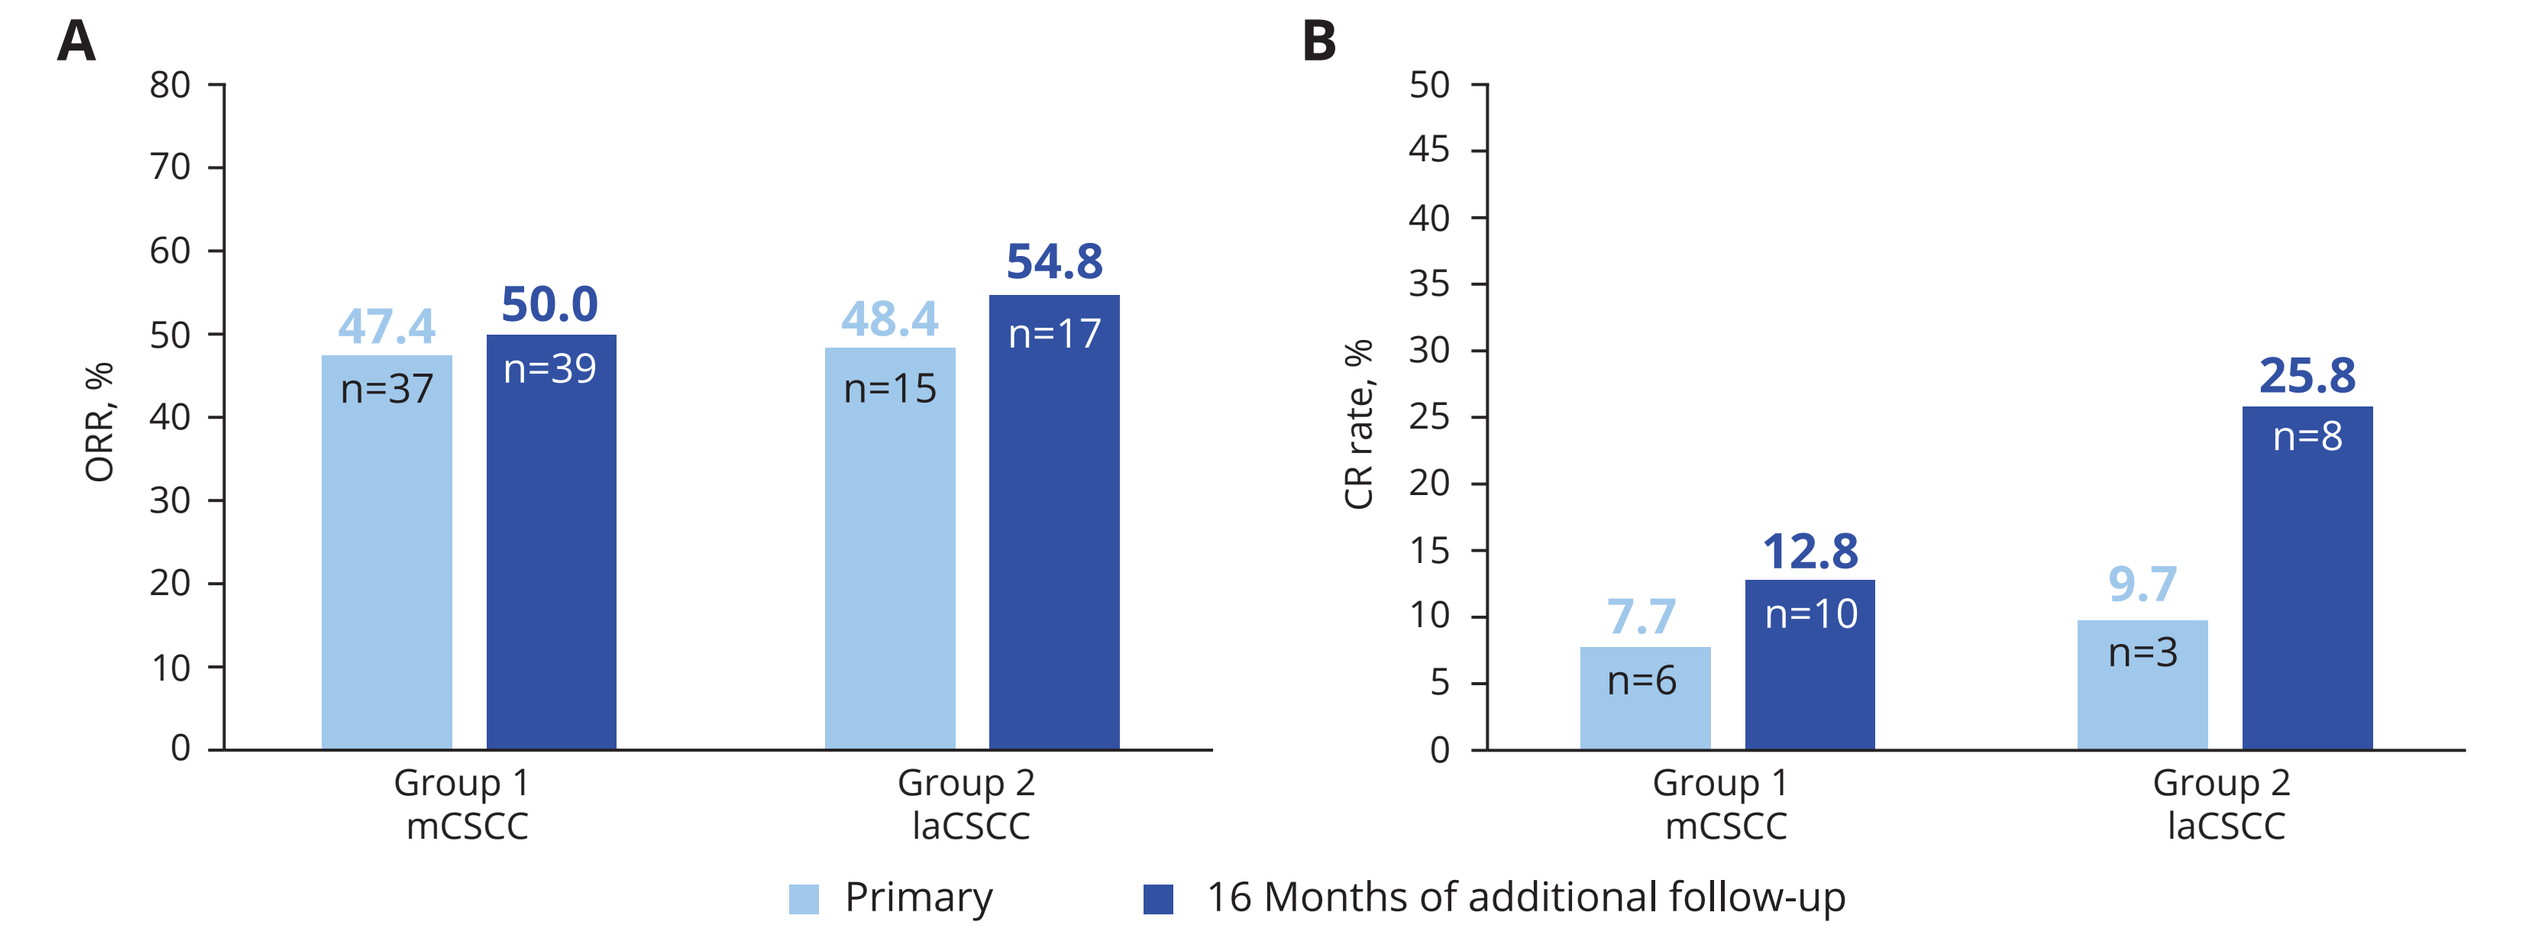

A
ORR, %
47.4
n=37
50.0
n=39
48.4
n=15
54.8
n=17
Group 1 mCSCC
Group 2 laCSCC
B
CR rate, %
7.7
n=6
12.8
n=10
9.7
n=3
25.8
n=8
Group 1 mCSCC
Group 2 laCSCC
Primary
16 Months of additional follow-up

CR, complete response; ICR, independent central review; laCSCC, locally advanced cutaneous squamous cell carcinoma; mCSCC, metastatic cutaneous squamous cell carcinoma; ORR, objective response rate.

Figure 4. Kaplan-Meier curves of DOR per ICR.[a]

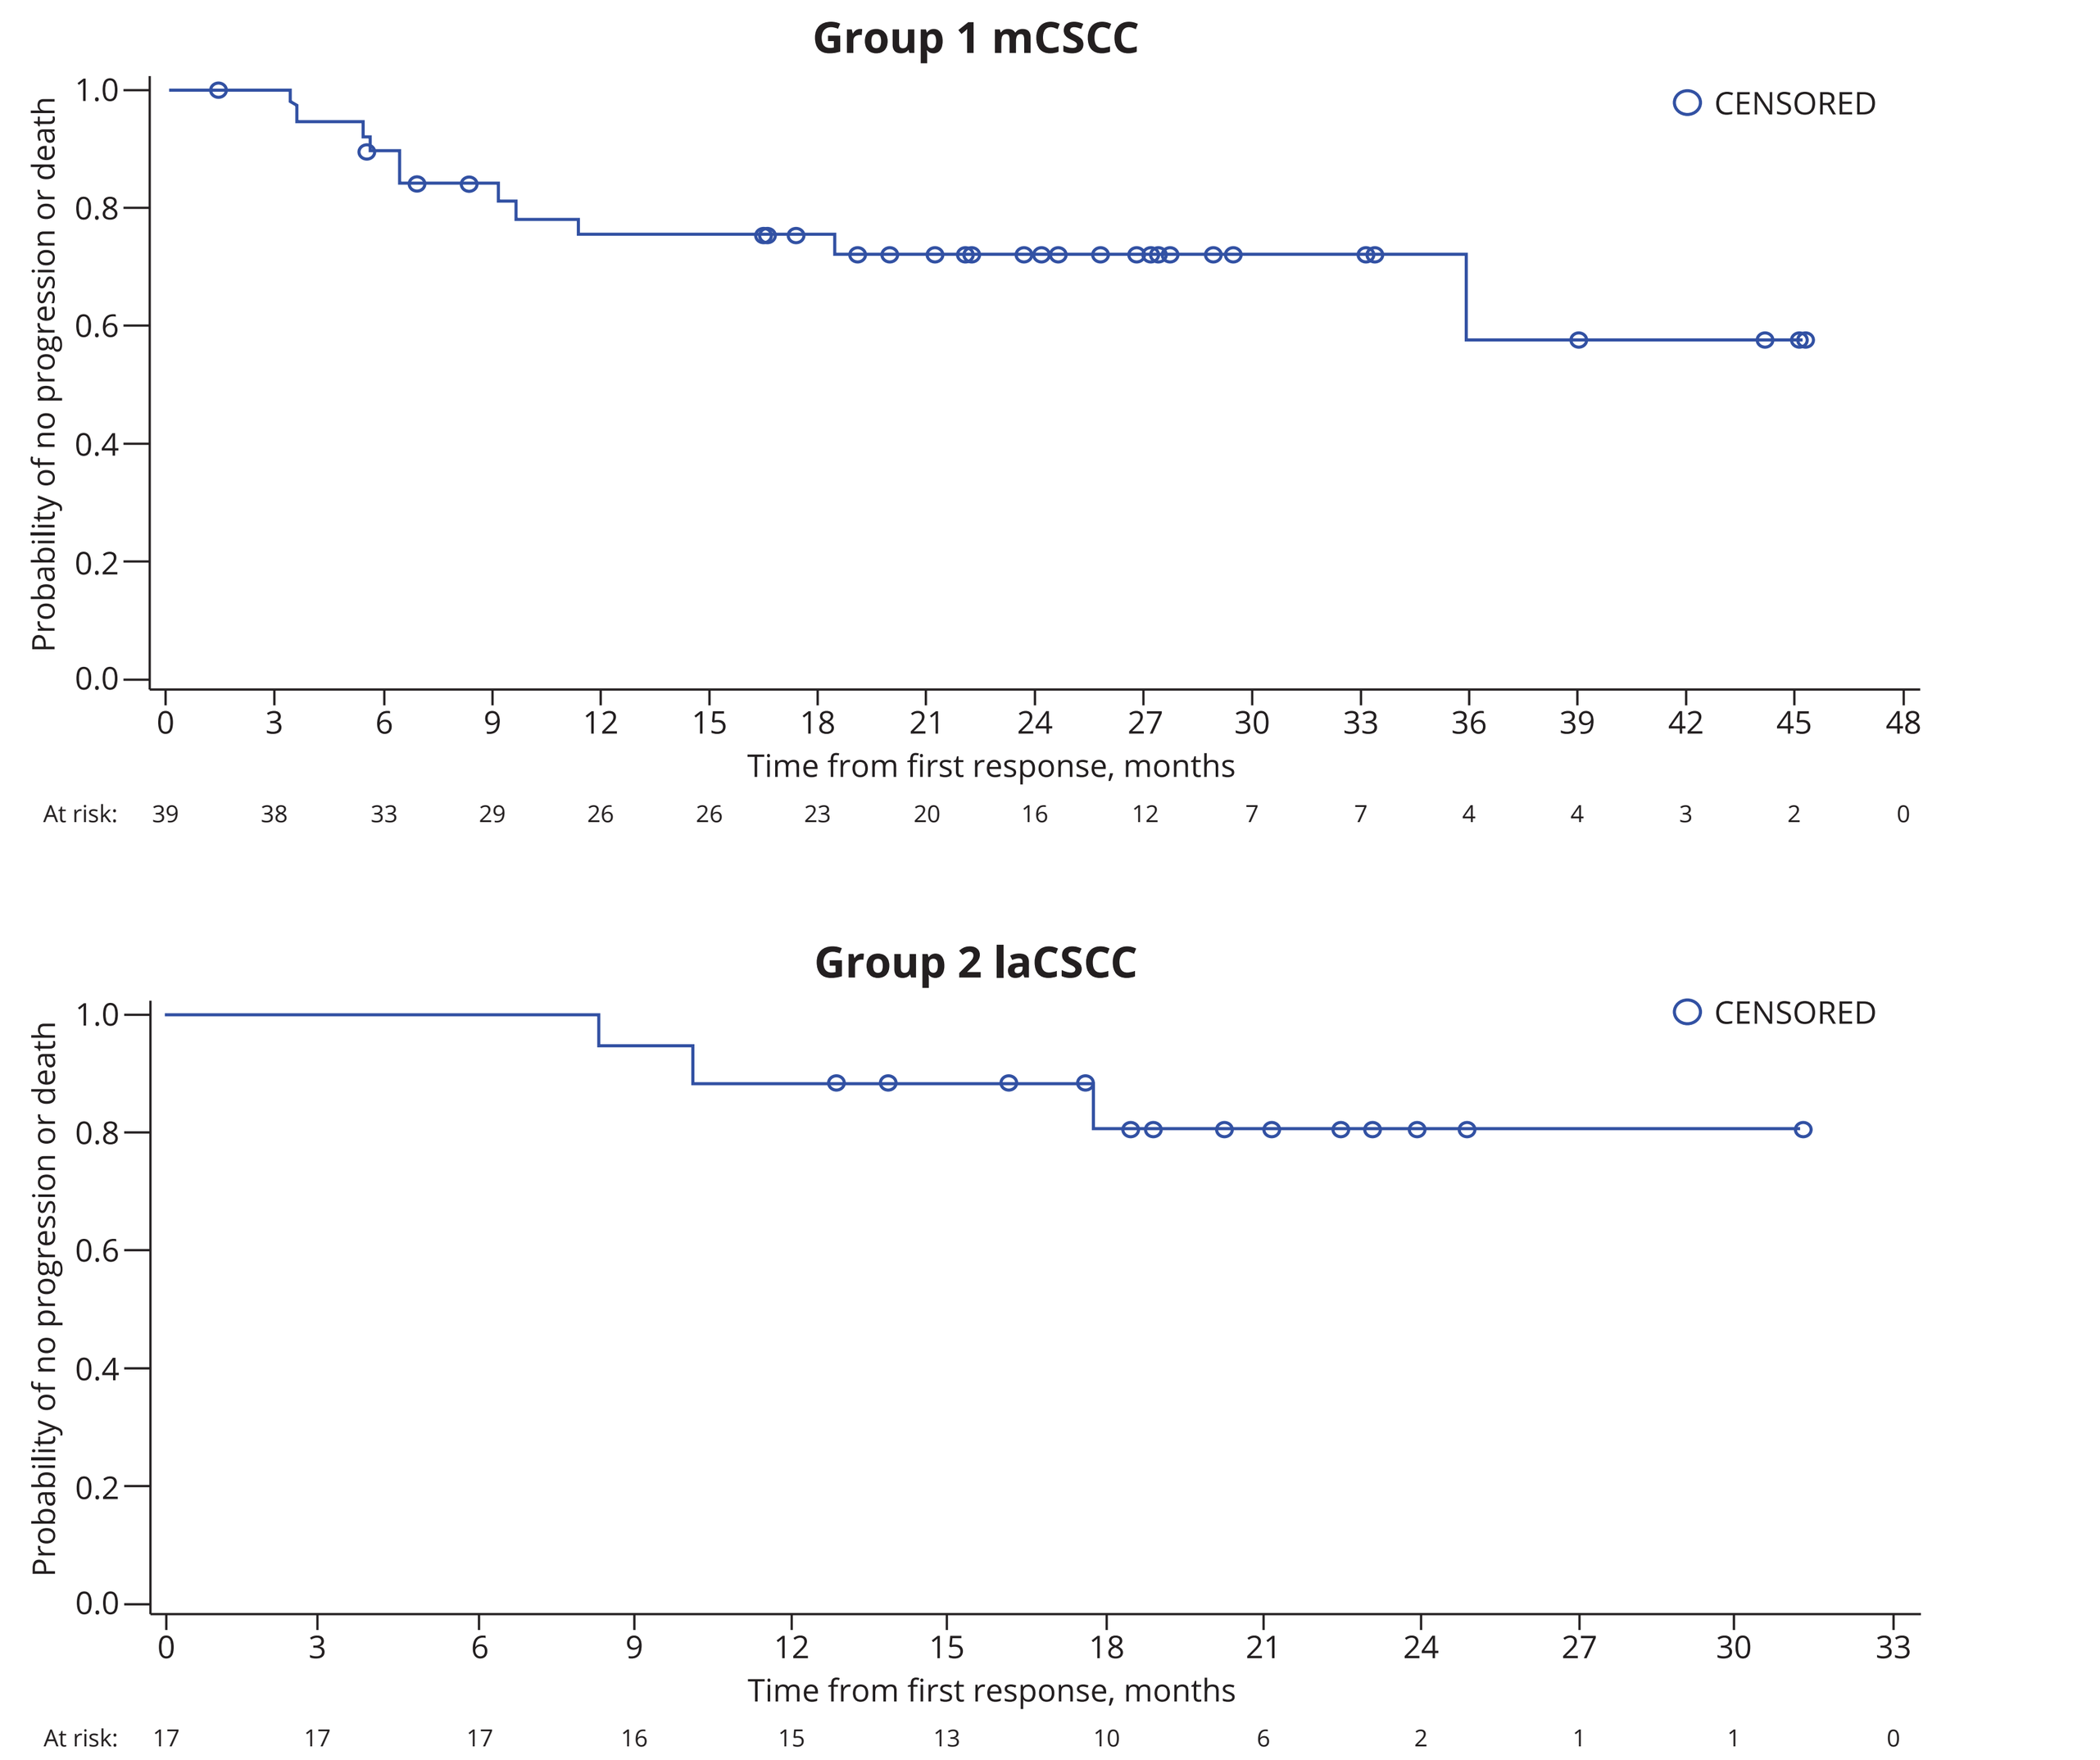

Group 1 mCSCC
CENSORED
Probability of no progression or death
Time from first response, months
At risk: 39 38 33 29 26 26 23 20 16 12 7 7 4 4 3 2 0
Group 2 laCSCC
CENSORED
Probability of no progression or death
Time from first response, months
At risk: 17 17 17 16 15 13 10 6 2 1 1 0

CR, complete response; DOR, duration of response; ICR, independent central review; laCSCC, locally advanced cutaneous squamous cell carcinoma; mCSCC, metastatic cutaneous squamous cell carcinoma; PR, partial response. [a]DOR is measured from the time measurement criteria are first met for response (CR or PR, whichever is first recorded) until the first date of recurrent or progressive disease (radiographic), or death due to any cause.

Safety

- Treatment-emergent adverse events (TEAEs), regardless of attribution, were reported in 181 patients (94.3%; Table 4)
 - The most common TEAEs of any grade were fatigue (22.9%), anemia (20.3%), constipation (16.1%), and diarrhea (15.1%)
 - The most common treatment-related TEAEs of any grade were pruritus and fatigue (each, 12.0%) and rash (8.9%)
- Fifty-three patients (27.6%) experienced immune-related adverse events (irAEs)
 - 3.6% assessed as grade 3, with no grade ≥4 irAEs
 - No events of grade ≥3 pneumonitis, colitis, hepatitis, nephritis, or endocrinopathies
- Permanent treatment discontinuation due to TEAEs, regardless of attribution, was observed in 12 patients (6.3%); the most common reason was COVID-19/COVID-19 pneumonia (1.6%)
- TEAEs led to death in 6 patients (3.1%), all considered unrelated to cosibelimab treatment
- The safety profile of cosibelimab was generally similar across groups and dosing regimens

Table 4. Summary of TEAEs Regardless of Attribution

	Advanced CSCC (n=192)	
TEAE, n (%)	**Any grade**	**Grade ≥3**
Any	181 (94.3)	87 (45.3)
Immune-related TEAE	53 (27.6)	7 (3.6)
Most common TEAEs (≥10%)[a]		
Fatigue	44 (22.9)	4 (2.1)
Anemia	39 (20.3)	10 (5.2)
Constipation	31 (16.1)	1 (0.5)
Diarrhea	29 (15.1)	—
Pruritus	26 (13.5)	—
Nausea	25 (13.0)	—
Rash	24 (12.5)	1 (0.5)
Asthenia	22 (11.5)	1 (0.5)
Arthralgia	21 (10.9)	—
Skin lesion	20 (10.4)	—
Headache	20 (10.4)	—

CSCC, cutaneous squamous cell carcinoma; TEAE, treatment-emergent adverse event. [a]TEAEs reported in ≥10% of patients, ordered by frequency of any grade.

FUNDING: This study was sponsored by Checkpoint Therapeutics.

ACKNOWLEDGMENTS: Writing and editorial support were provided under the direction of the authors by Ella A. Kasanga, PhD, PMP®, and Erin Reineck, ELS, of MedThink SciCom, and funded by Checkpoint Therapeutics.

REFERENCES: 1. Stanganelli I, Spagnolo F, Argenziano G, et al. *Cancers (Basel)*. 2022;14(2):377. **2.** Tokez S, Hollestein L, Louwman M, Nijsten T, Wakkee M. *JAMA Dermatol.* 2020;156(12):1300-1306. **3.** Palyca P, Koshenkov VP, Mehnert JM. *Am Soc Clin Oncol Educ Book*. 2014:e397-e404. **4.** Jones OT, Ranmuthu CKI, Hall PN, Funston G, Walter FM. *Adv Ther*. 2020;37(1):603-616. **5.** Perera E, Gnaneswaran N, Staines C, Win AK, Sinclair R. *Australas J Dermatol*. 2015;56(4):258-267. **6.** Twomey JD, Zhang B. *AAPS J*. 2021;23(2):39. **7.** Gorelik L, Avgerinos G, Kunes Y, Marasco WA. Poster presented at: The American Association for Cancer Research Annual Meeting; 1-5 April 2017; Washington, DC. **8.** Lin L, Hilbert J, Gorelik L, et al. Abstract presented at: The Society for Immunotherapy of Cancer 34th Annual Meeting; 6-10 November 2019; National Harbor, MD. **9.** Plock N, Kleijn HJ, Neighbours L, Oliviero J. Poster presented at: The Population Approach Group Europe Annual Meeting; 27-30 June 2023; A Coruña, Spain. **10.** US Food and Drug Administration. https://www.fda.gov/media/151745/download. Accessed 22 August 2024.